PROGRAM # 1068.27

Abstract # 5794

Inverse relationship between innate and acquired immune responsiveness in mice initially dosed with cationic lipid DNA complexes (JVRS-100)

Marla Lay[1], Bernadette Callejo[1], Thierry Giffon[2], David B. Lewis[2], Stella Chang[1], Jeffery Fairman[1]

[1]Juvaris BioTherapeutics, Pleasanton, CA, [2]Department of Pediatrics and the Program in Immunology, Stanford University School of Medicine, Stanford, CA

JUVARIS BIOTHERAPEUTICS, INC.

ABSTRACT

Hyporesponsiveness to excessive innate immune stimulation (i.e., lipopolysaccharide, lipoteichoic acid, and CpG oligonucleotides) has been studied extensively in vitro and documented in clinical treatment of sepsis patients with low inflammatory cytokine production, reduced leukocyte expression of HLA-DR, and decreased antigen presentation. Closely spaced intravenous (IV) administration of JVRS-100 has been shown to induce a short (7-10 day) refractory period for innate immune activation, with 50% of responsiveness restored after three days and 100% by 10-14 days. This refractory period was observed in interferon-gamma and IFN-gamma receptor knockout mice as well as mice pre-dosed with IFN-gamma or depleted of NK cells or plasmacytoid dendritic cells. In contrast to reduced capacity for antigen presentation (sepsis patients), IV administration of JVRS-100, one day prior to vaccination with HKx31 heat-inactivated influenza virus adjuvanted with JVRS-100 significantly enhanced the influenza-specific humoral immune response. Enhancement of the adaptive response to vaccination was greatest at the timepoints showing the maximum refractory period, suggesting an inverse relationship between cytokine production and immune activation. These findings may have implications for development of future vaccines, in which single or limited vaccination strategies are critical.

This work was supported by NIAID grant “Innate Immune Stimulation as a Pathogen Countermeasure” 1R43AI060146-01A2

RESULTS

Refractory Period to Multiple IV Doses of JVRS-100 in Mice

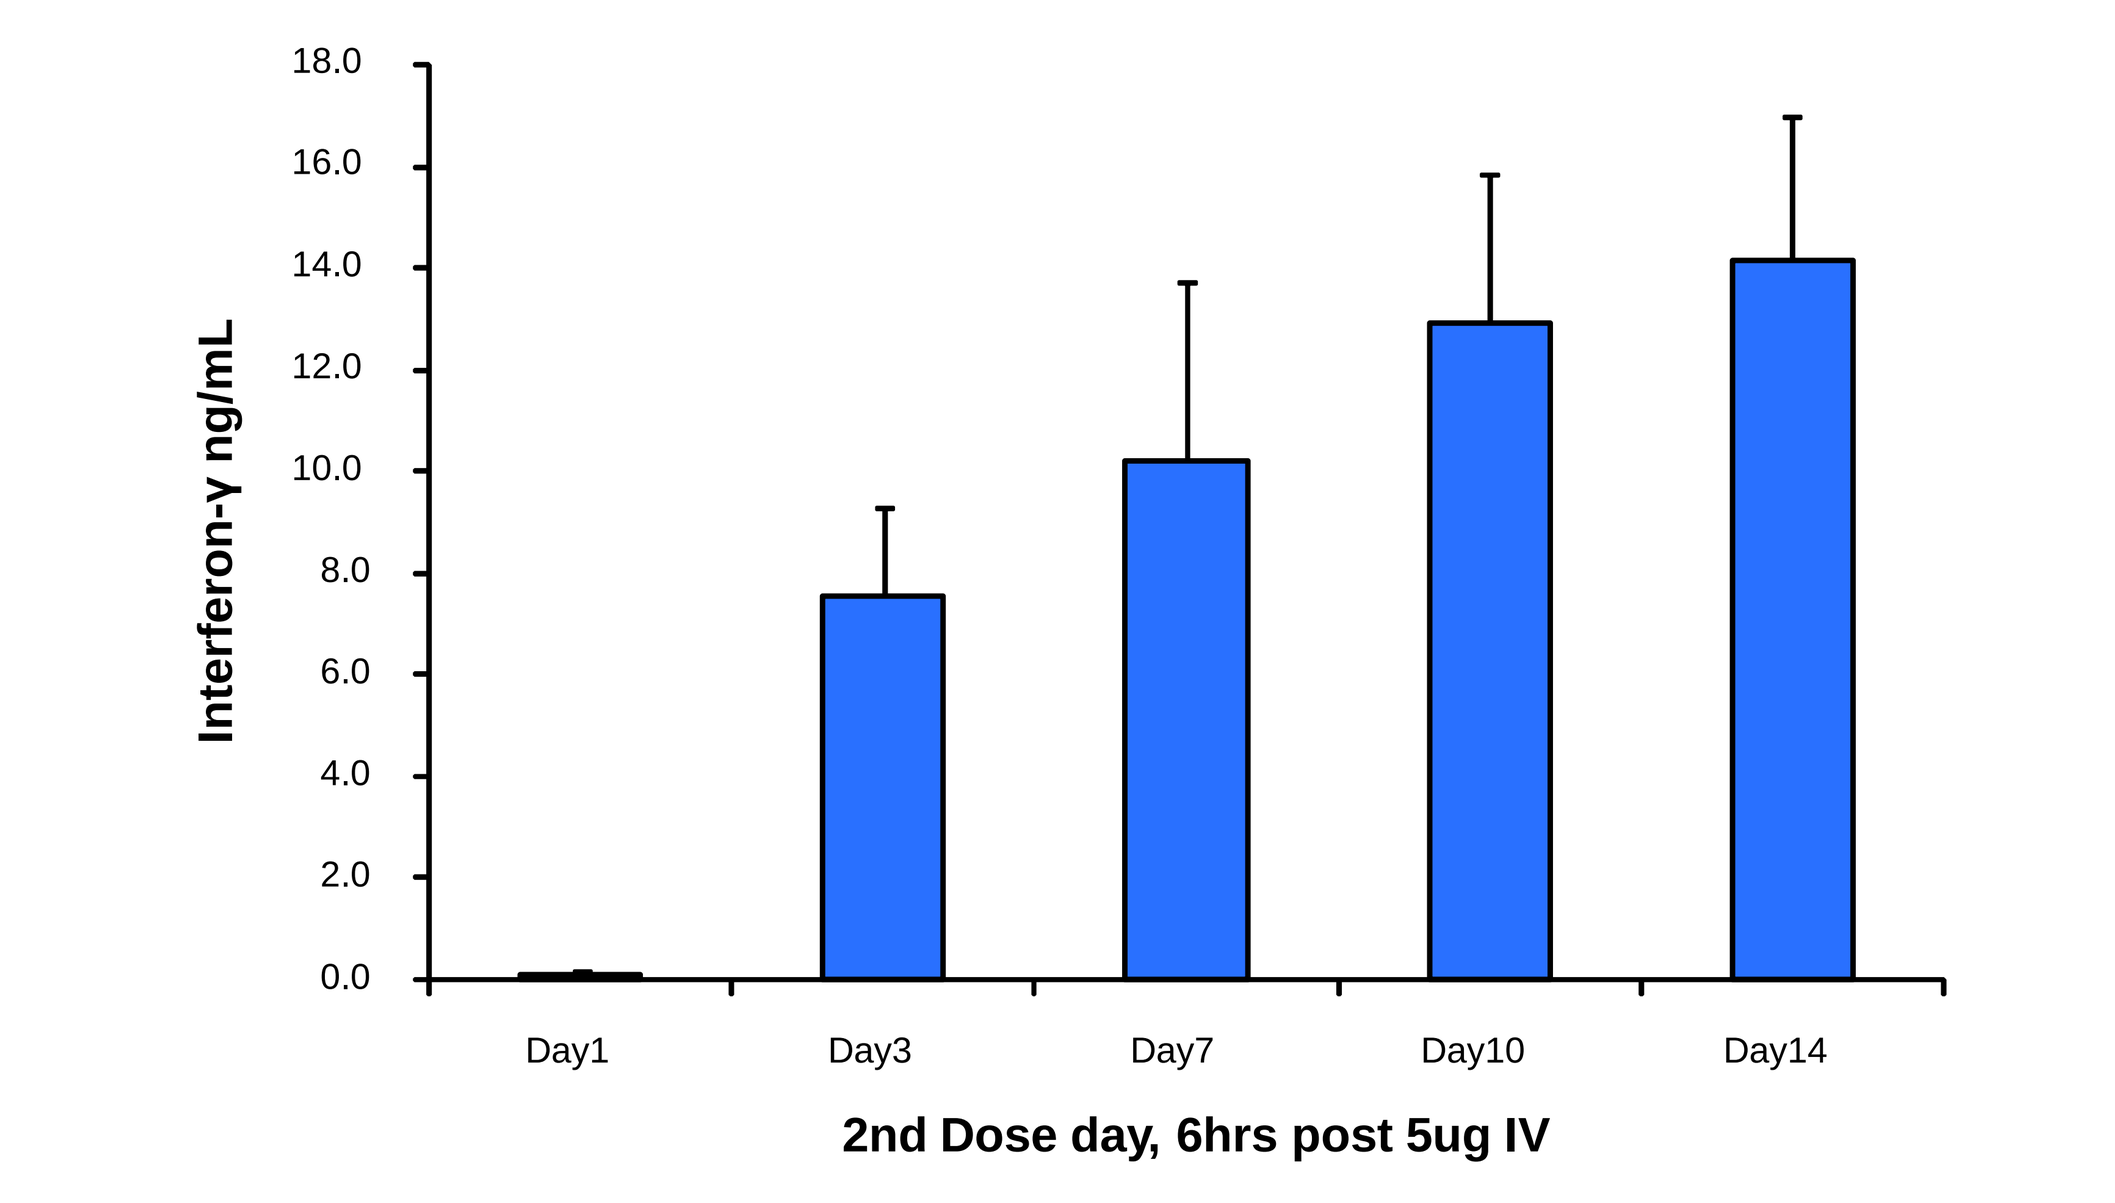
Interferon-γ ng/mL
18.0
16.0
14.0
12.0
10.0
8.0
6.0
4.0
2.0
0.0
Day1
Day3
Day7
Day10
Day14
2nd Dose day, 6hrs post 5ug IV

Comparable results with 10ug IV (duration non-dose dependent)

CD-1 mice were intravenously administered 5µg JVRS-100 on day 0 followed by a second IV 5µg dose on the specified day. Innate immune activation was measured by ELISA assay for serum Interferon-ɤ.

Refractory Period to Multiple IV Doses of JVRS-100 in Mice is Dose Dependent in Magnitude

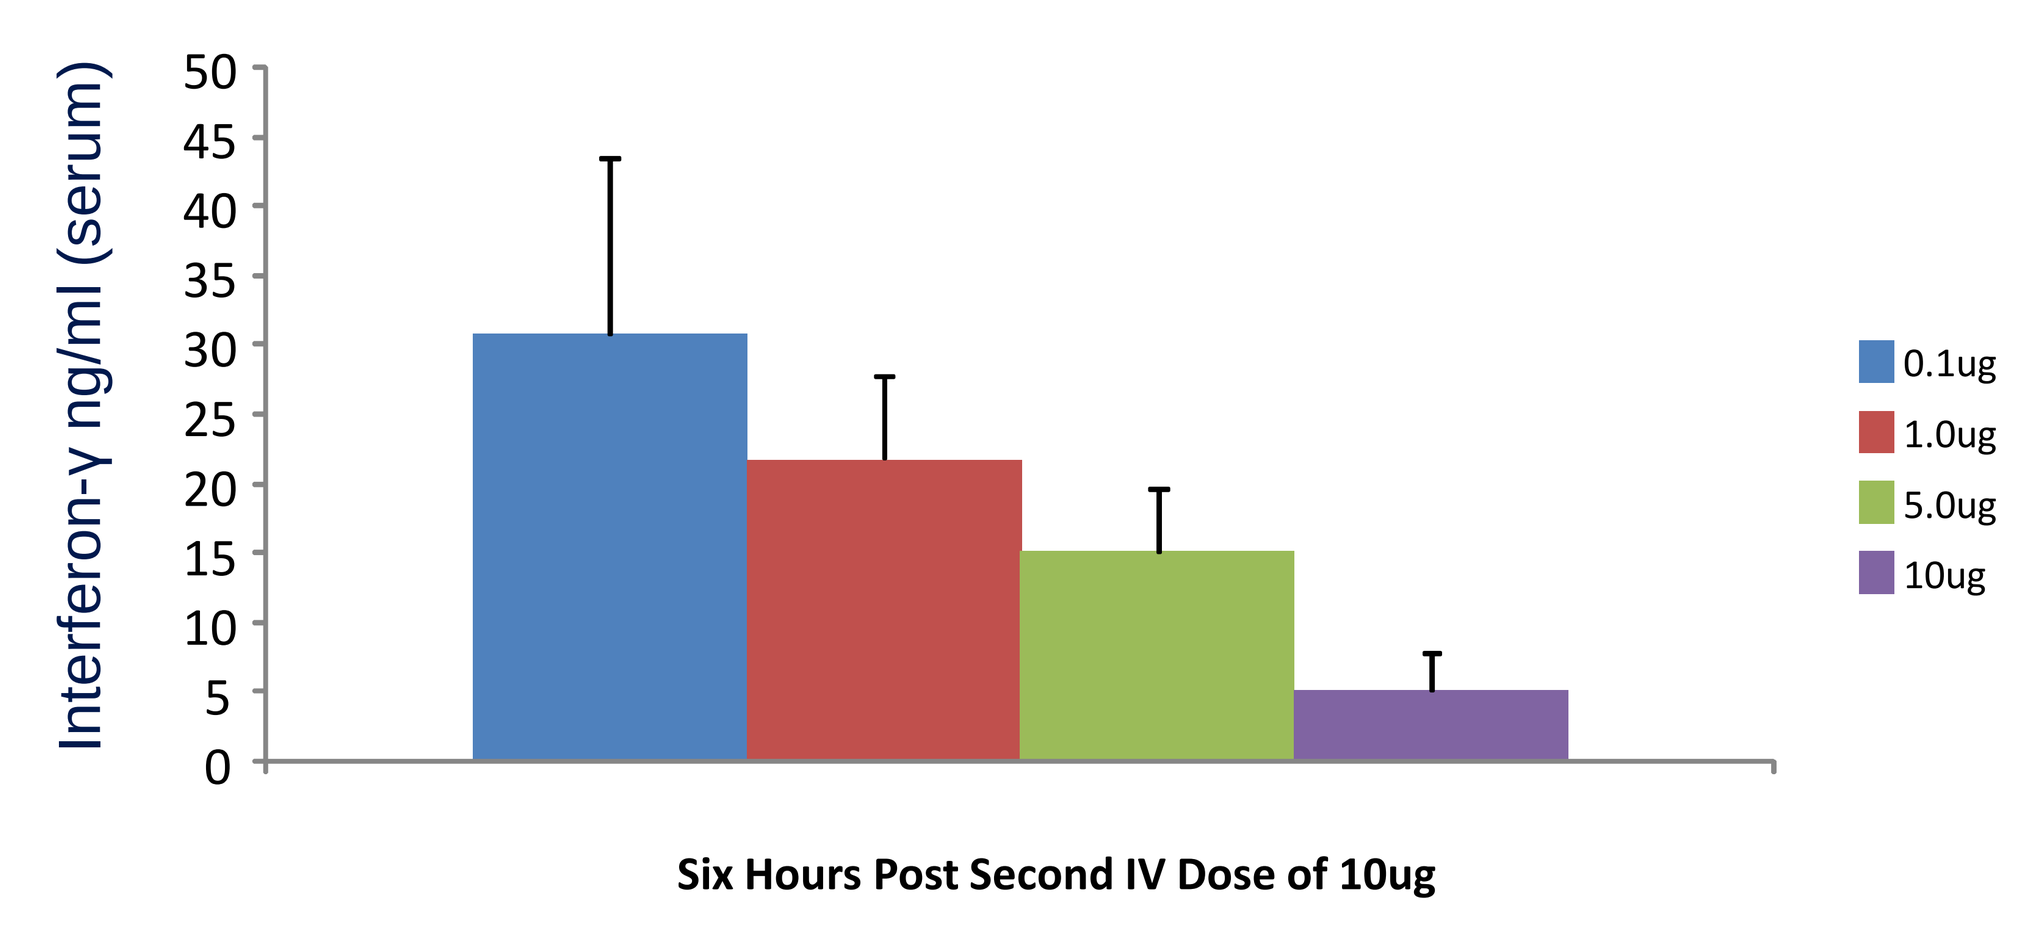
Interferon-γ ng/ml (serum)
50
45
40
35
30
25
20
15
10
5
0
0.1ug
1.0ug
5.0ug
10ug
Six Hours Post Second IV Dose of 10ug

CD-1 mice were intravenously administered one of four increasing doses of JVRS-100 on day 0 followed by a second IV 10µg dose on day 1. Innate immune activation was measured by ELISA assay for serum Interferon-ɤ.

Refractory Period to Multiple Doses is Not Dependent on Interferon-Gamma Signaling

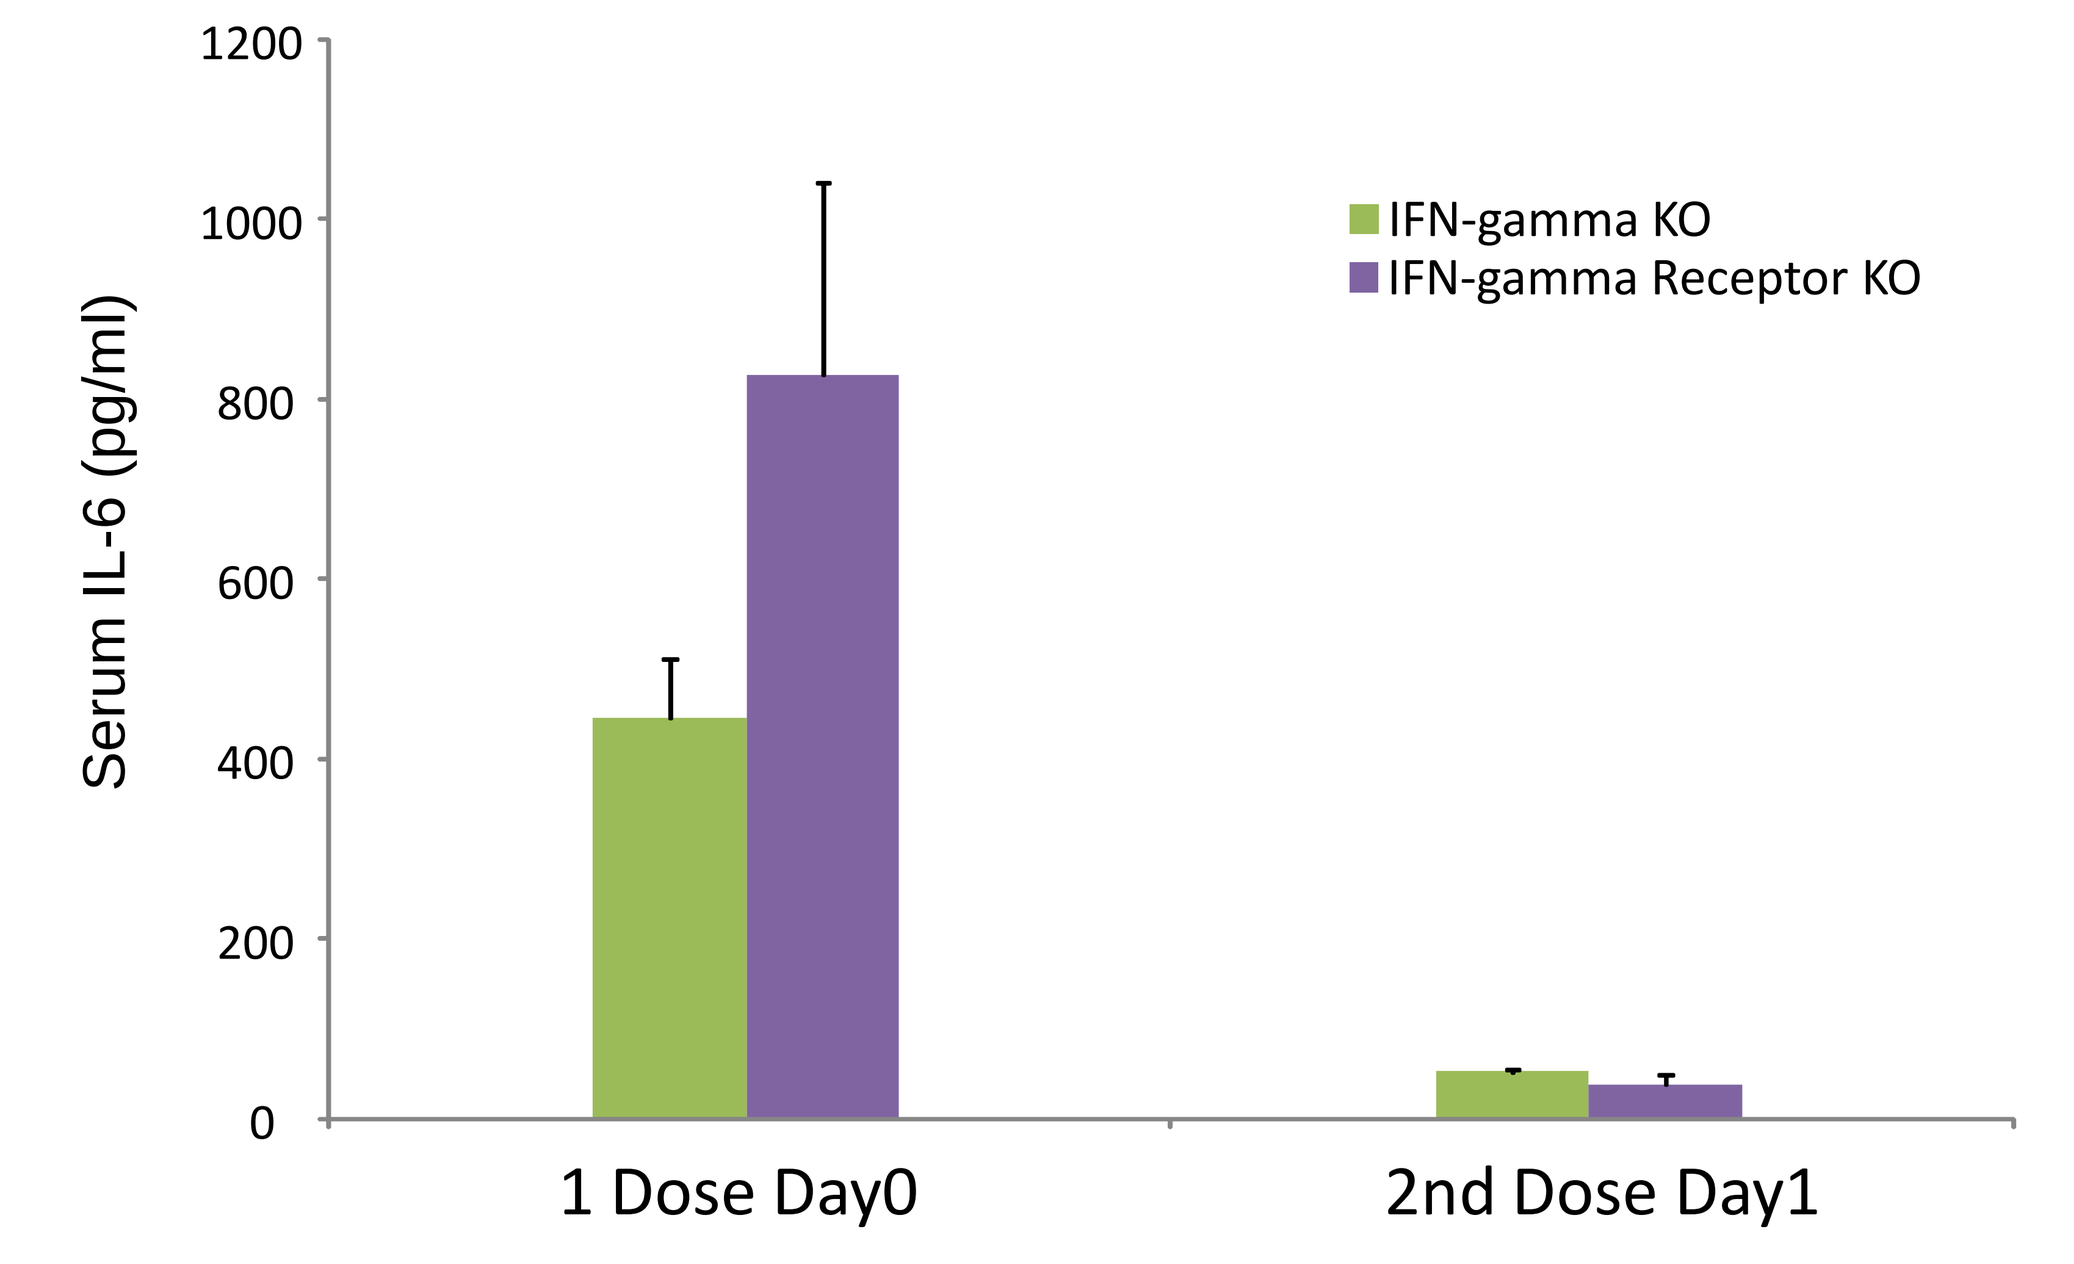
Serum IL-6 (pg/ml)
1200
1000
800
600
400
200
0
IFN-gamma KO
IFN-gamma Receptor KO
1 Dose Day0
2nd Dose Day1

Specified knockout mice were intravenously administered 5µg JVRS-100 on day 0 followed by a second IV dose one day later. Innate immune activation was measured by ELISA assay for serum IL-6.

Enhanced Vaccine Immunogenicity as Measured by HAI Response Following HKx31 Vaccination

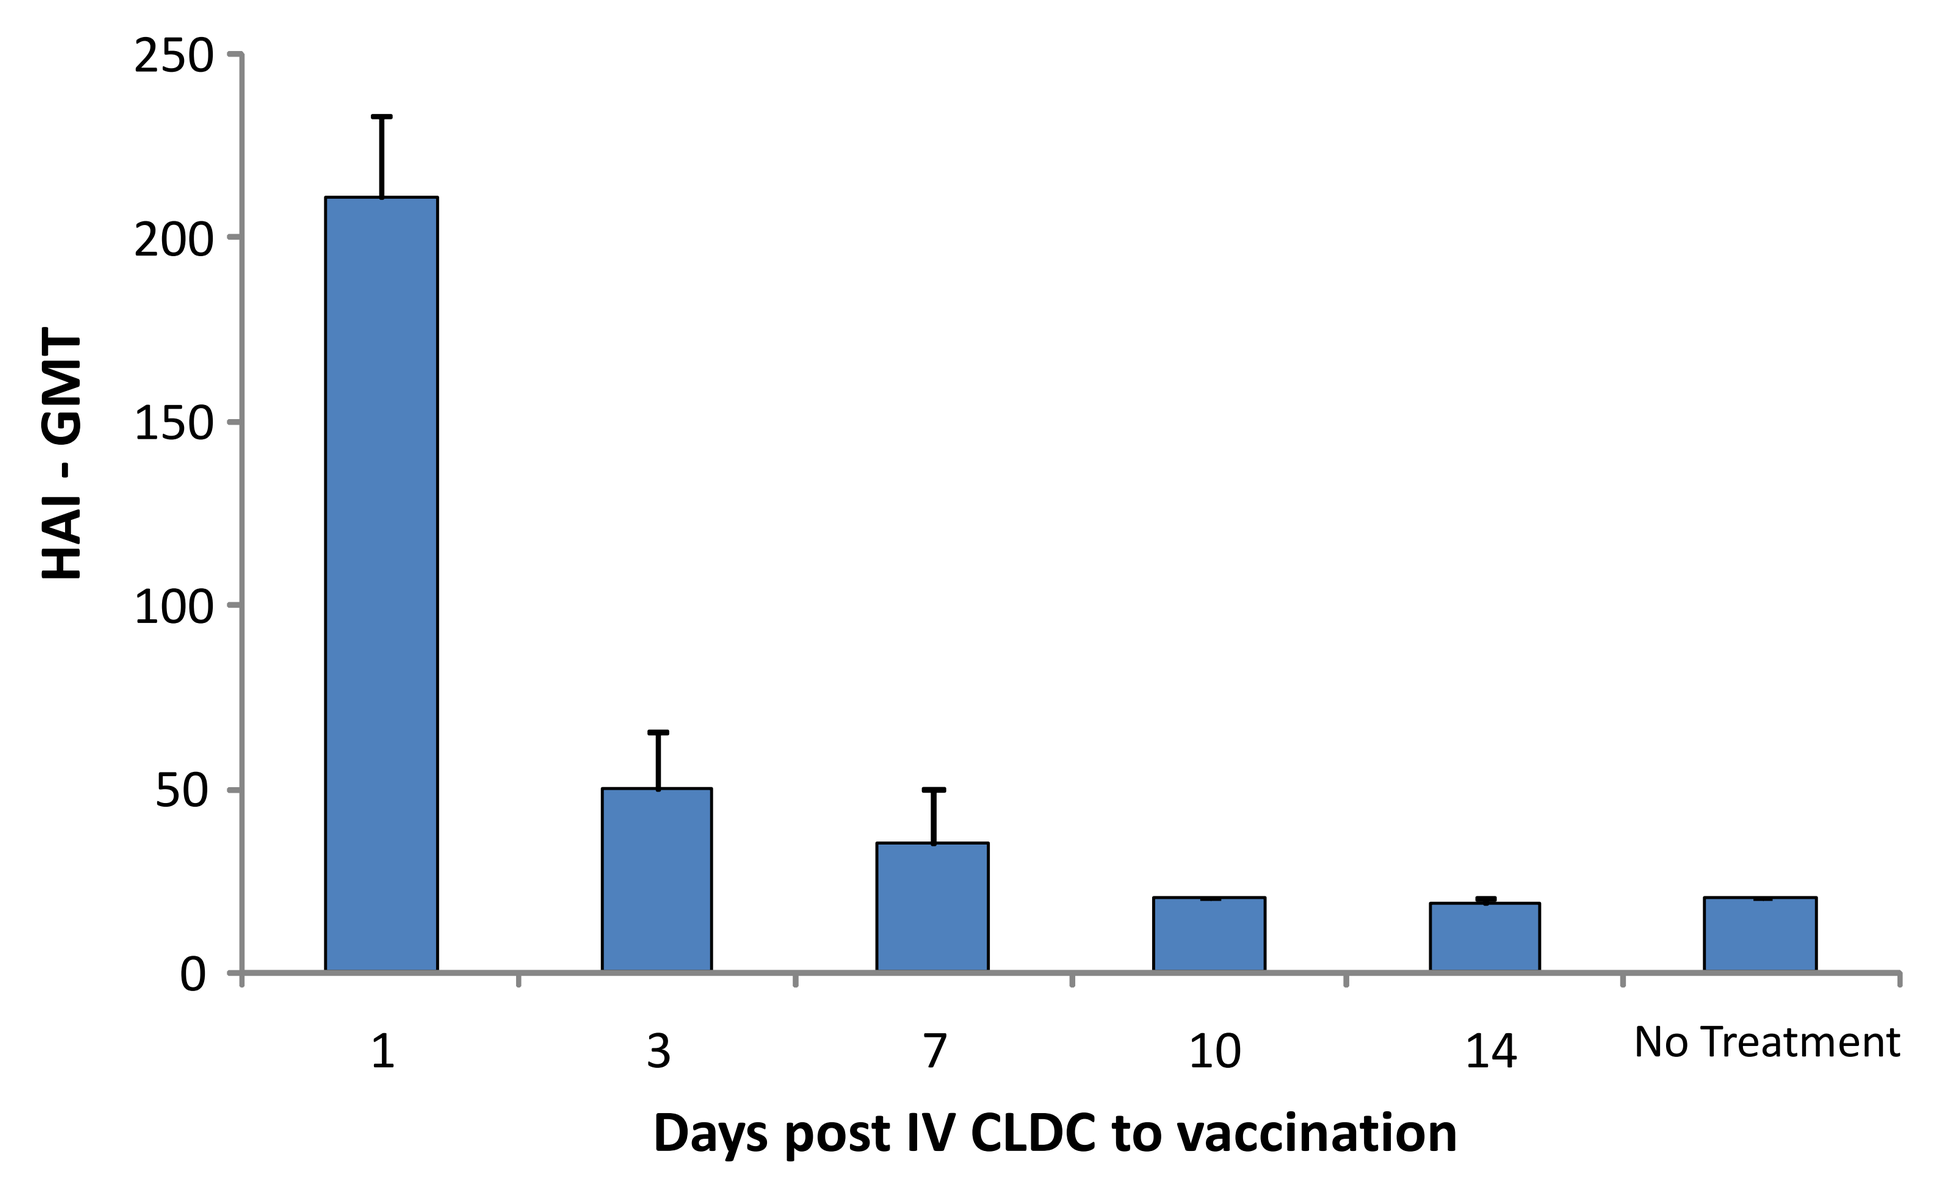
HAI - GMT
250
200
150
100
50
0
1
3
7
10
14
No Treatment
Days post IV CLDC to vaccination

CD-1 mice were intravenously administered 5µg JVRS-100 on day 0 followed by a subcutaneous vaccination of 5ug heat-inactivated HKx31 influenza virus on the specified day. Immunogenicity was measured by a hemagglutination inhibition titer.

Cellular Response as Measured by Interferon-Gamma Response to Restimulation with Antigen

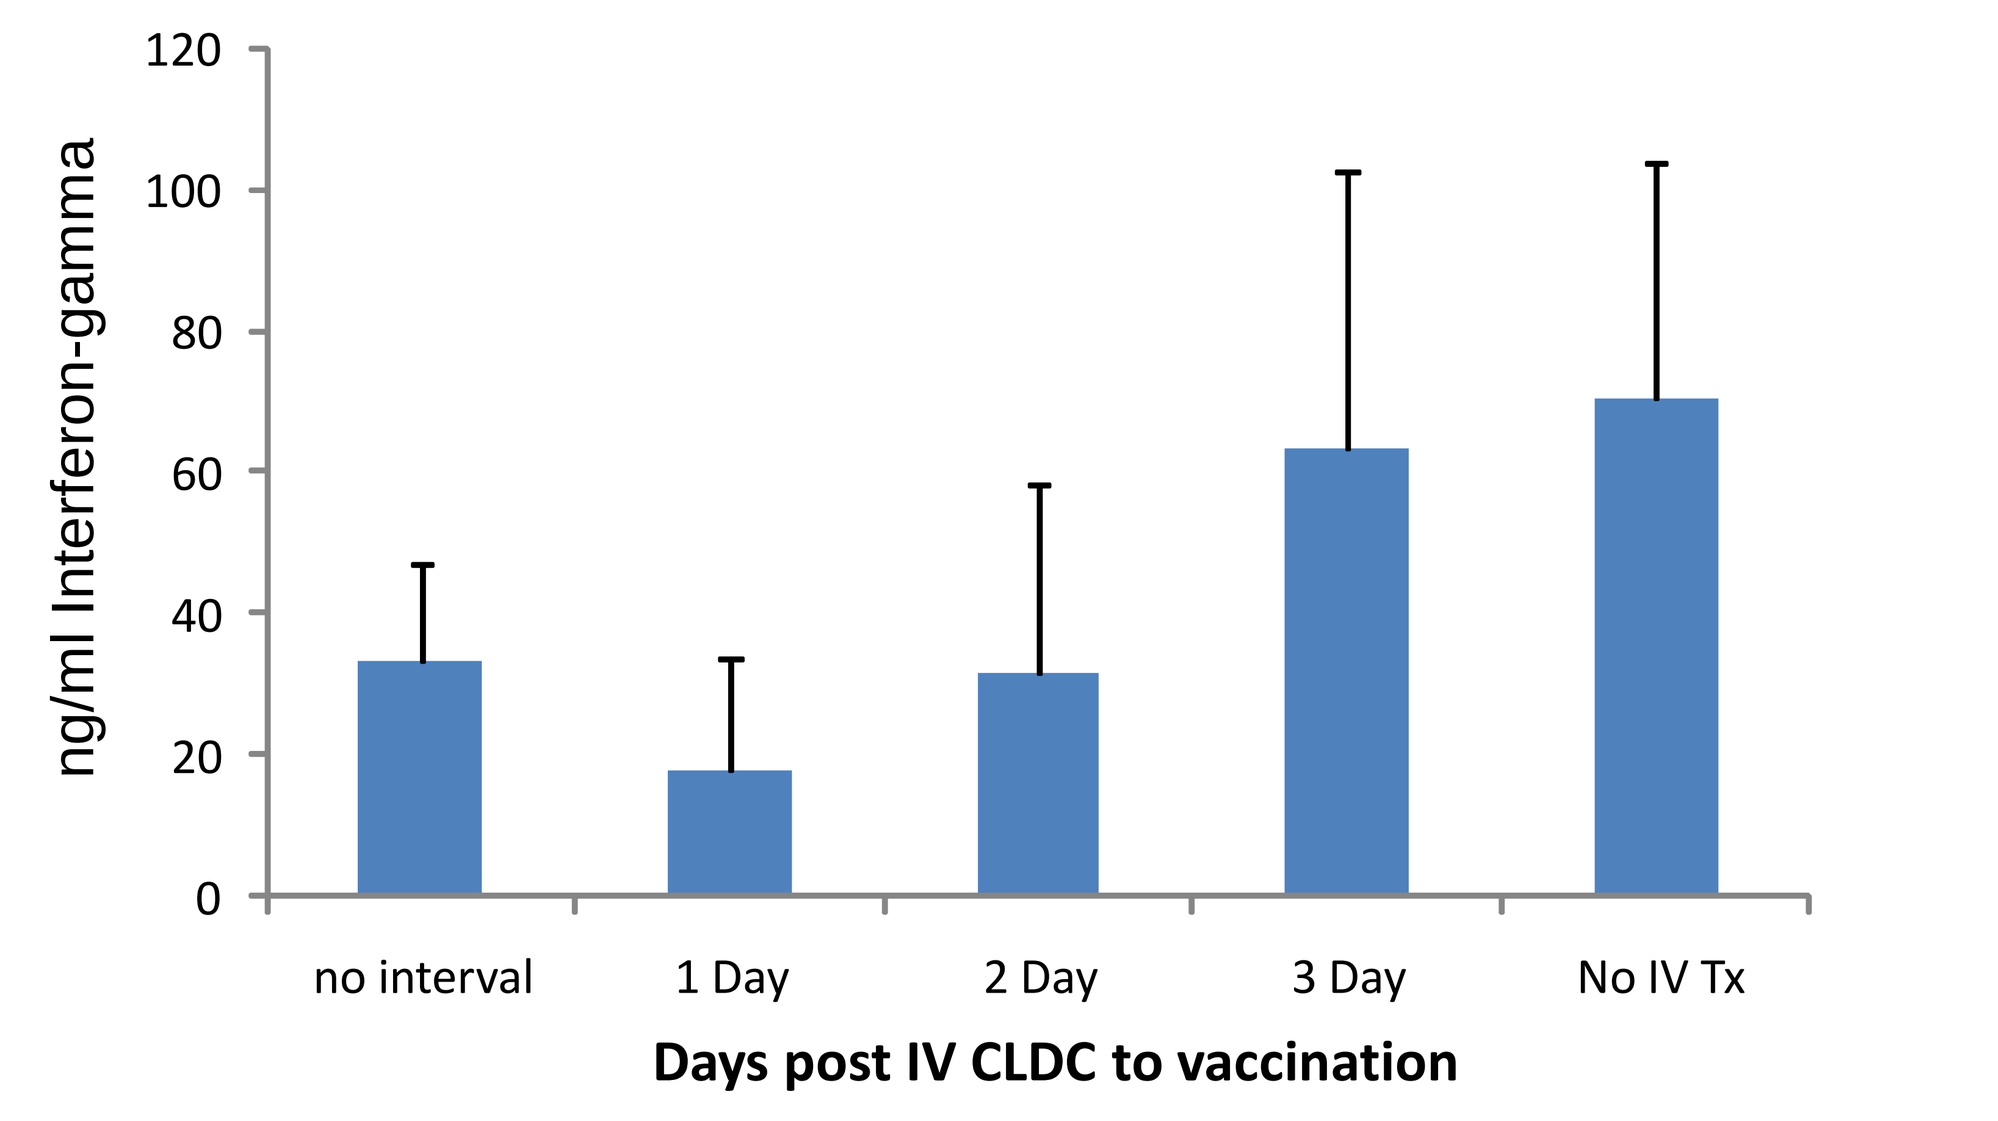
ng/ml Interferon-gamma
120
100
80
60
40
20
0
no interval
1 Day
2 Day
3 Day
No IV Tx
Days post IV CLDC to vaccination

Splenocytes from vaccinated mice were restimulated with HKx31 at 1ug/ml

CD-1 mice were administered 5µg JVRS-100 IV on day 0 followed by a SC vaccination of 5ug heat-inactivated HKx31 influenza virus on the specified day. Cellular response was estimated by ELISA assay for interferon-ɤ in an antigen stimulated splenocyte culture.

Enhanced Vaccine Immunogenicity as Measured by HAI Response Following Fluzone® Vaccination

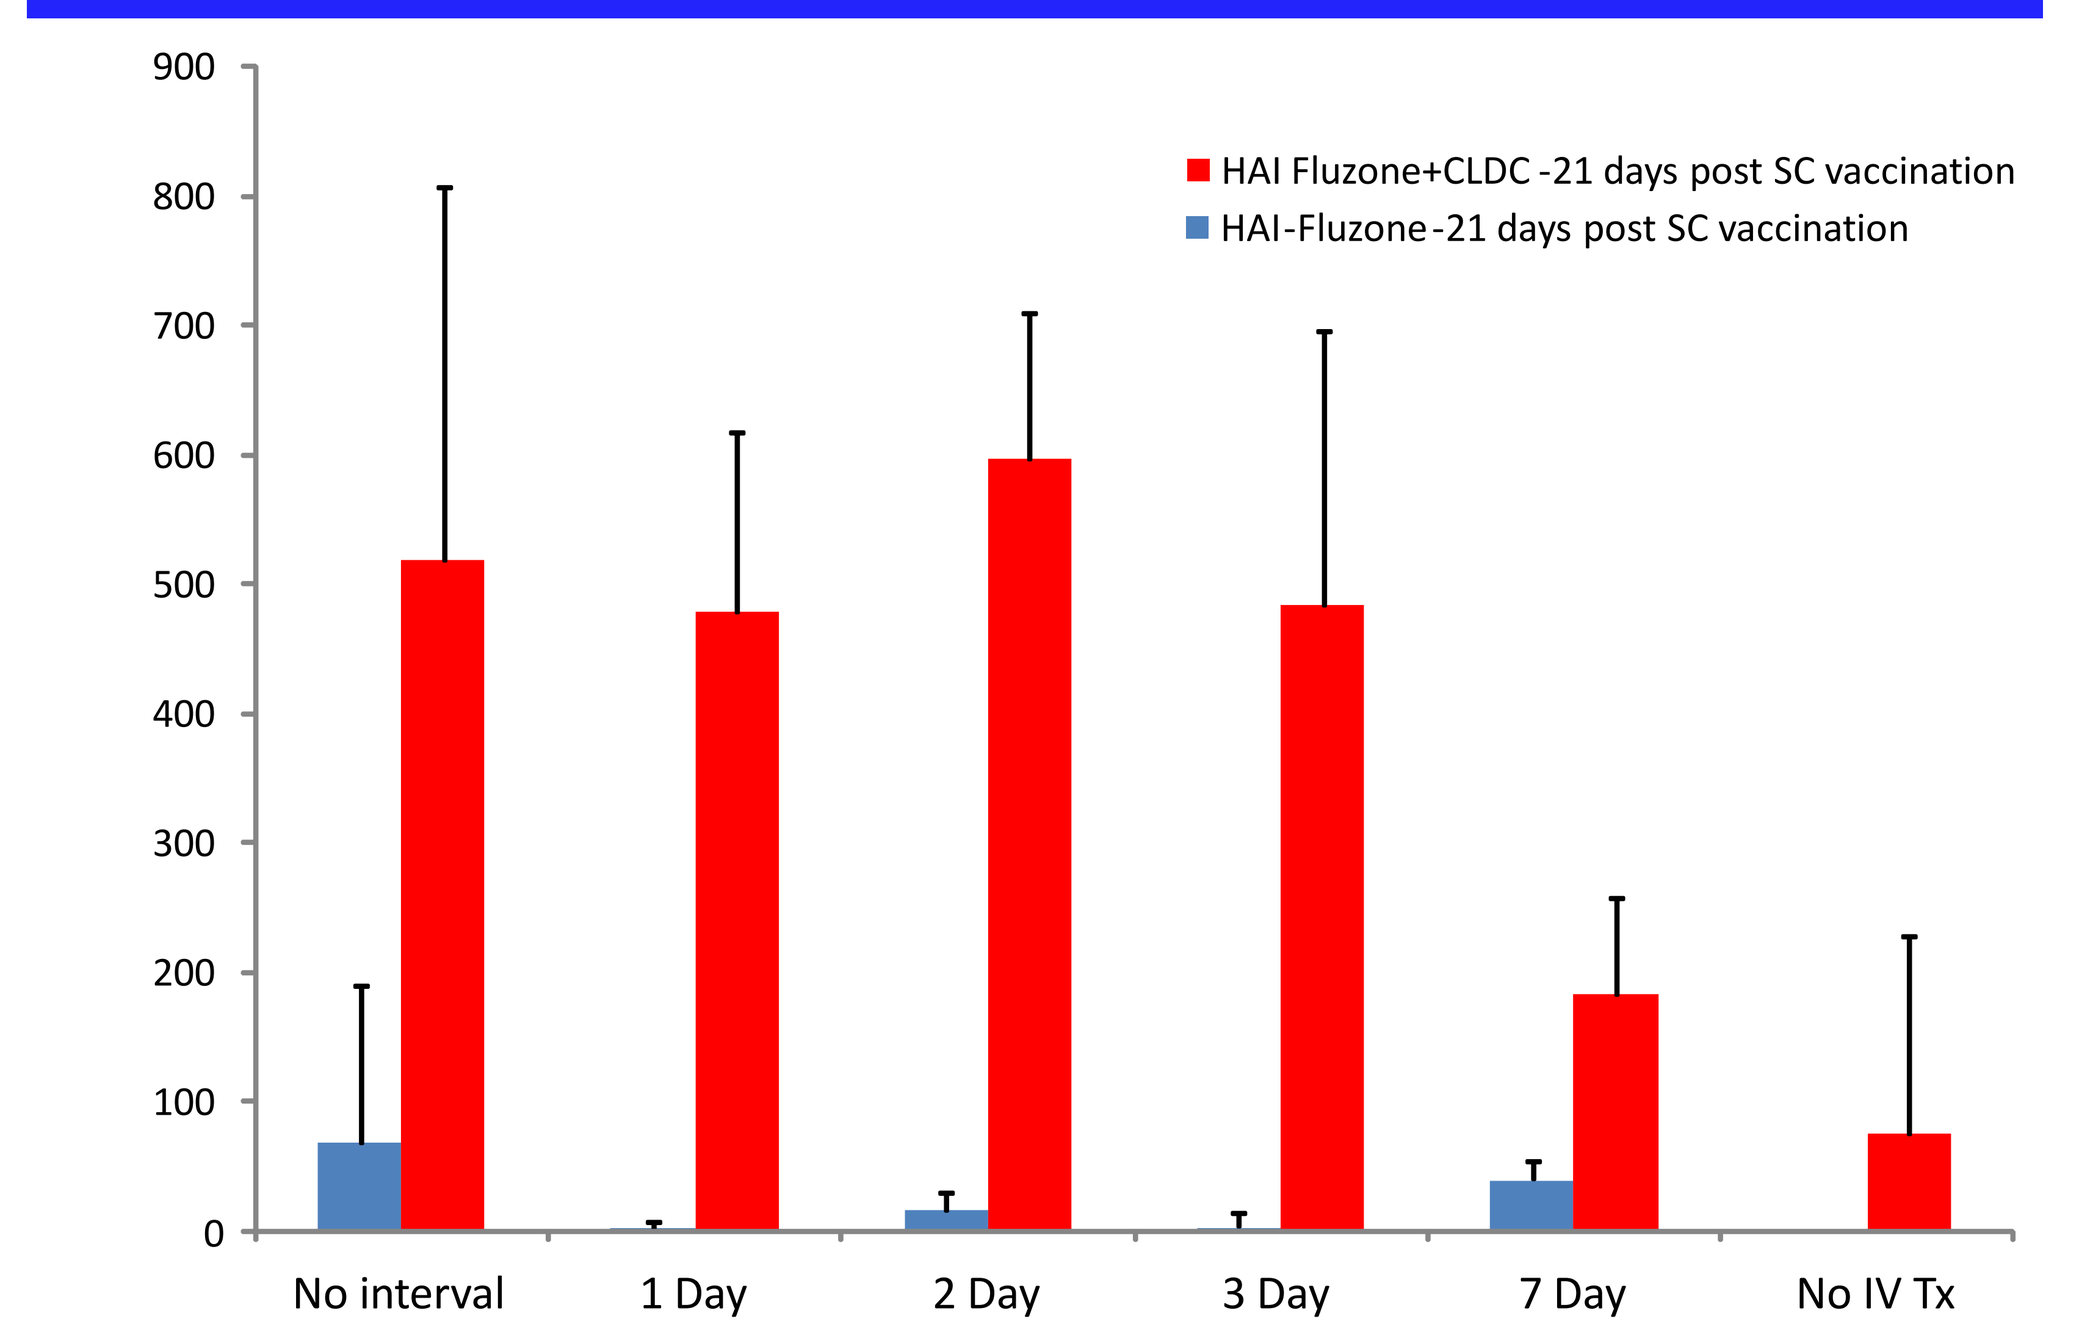
900
800
700
600
500
400
300
200
100
0
HAI Fluzone+CLDC -21 days post SC vaccination
HAI-Fluzone -21 days post SC vaccination
No interval
1 Day
2 Day
3 Day
7 Day
No IV Tx

CD-1 mice were intravenously administered 5µg JVRS-100 on day 0 followed by a subcutaneous vaccination of 5ug Fluzone® on the specified day. Immunogenicity was measured by a hemagglutination inhibition titer.

CONCLUSIONS

These studies demonstrate that there is a period of hyporesponsiveness to IV administration of JVRS-100, similar to other immune stimulants that is dose-dependent in magnitude, but not in duration. Furthermore, systemic immune stimulation using JVRS-100 alone can enhance the antibody response to subsequent parenteral vaccination using JVRS-100 adjuvanted vaccines. In contrast, there appears to be little impact on the cellular immune response, although the authors hypothesize that the CD4:CD8 ratio is altered following an IV JVRS-100 predose regimen. Experiments are underway to test this hypothesis. The observation has obvious implications in settings, in which a robust adaptive immune response is required from a single vaccination (i.e. therapeutic vaccination, pandemic flu, biodefense) and has the added benefit of eliciting a cytokine response that may be beneficial while the adaptive response to the vaccine antigen is initiated.

For More Information

For more information on this poster or the JVRS-100 immunostimulant and adjuvant please contact:

Jeff Fairman, Ph.D.
Co-Founder and Vice President, Research
Juvaris BioTherapeutics, Inc.
863A Mitten Road
Burlingame, CA 94010
(408) 857-4430
jfairman@juvaris.com